Preclinical evaluation of neratinib plus T-DM1 in orthotopic PDX models of HER2-positive breast cancer brain metastases

Jing Ni[1], Yanzhi Wang[1], Irmina Diala[2], Sheheryar Kabraji[1], Rachel A. Freedman[1], Nancy U. Lin[1], Jean J. Zhao[1].
[1]Dana-Farber Cancer Institute, Boston, MA. [2]Puma Biotechnology, Los Angeles, CA, USA.

Abstract

Up to half of patients with metastatic HER2-positive breast cancer will develop brain metastases. Breast cancer brain metastases (BCBM) are a major cause of morbidity and mortality, despite multimodal management including surgery, radiotherapy, and systemic therapies. Therefore, there is an urgent need to develop novel, efficacious alternatives. Neratinib is an orally bioavailable, irreversible pan-HER tyrosine kinase inhibitor that is FDA-approved in the extended adjuvant treatment setting for HER2-positive, early breast cancer (NCT00878709). Neratinib has only modest activity as a single agent in clinical trials of patients with HER2-positive brain metastases. Though the combination of neratinib and capecitabine results in CNS responses in up to half of patients, patients eventually develop drug resistance, and toxicities have been a concern (TBCRC022), thus exploration of alternative neratinib combinations is of significant clinical interest. Ado-trastuzumab emtansine (T-DM1) is an antibody-drug conjugate with reported single-agent activity against HER2-positive BCBM. Here, we used HER2-positive orthotopic patient derived xenograft (PDX) models of BCBM to test if combining neratinib with T-DM1 could improve tumor response. PDX cells are labelled with luciferase to allow tumor growth measurement *in vivo*. We found that neratinib is able to reduce phosphorylated HER2 in an orthotopic PDX tumor derived from HER2-positive BCBM, indicating that neratinib can cross the BBB and inhibit HER2 activation in BCBM PDX tissues. However, in both HER2-positive DF-BM354 and DF-BM355 PDX models, single agent neratinib did not block orthotopic tumor growth compared to vehicle control as monitored by bioluminescence measurements. In contrast, combined treatment of neratinib with T-DM1 significantly reduced tumor growth compared to single agent treatment with neratinib or T-DM1 at earlier time points in both models. At later time points, the combined treatment is comparable to T-DM1 alone in DF-BM354 model, but significantly prolong the survival of mice bearing DF-BM355 tumors. These data warrant further testing of neratinib alone and in combination with T-DM1 in additional BCBM PDX models to better understand drivers of resistance and susceptibility to HER2-inhibitors in HER2-positive BCBMs. Furthermore, they support the launch of a prospective clinical trial (NCT01494662) to test the efficacy and tolerability of T-DM1 in combination with neratinib in patients with progressive HER2-positive BCBM.

Results

Modeling metastatic breast cancer to brain

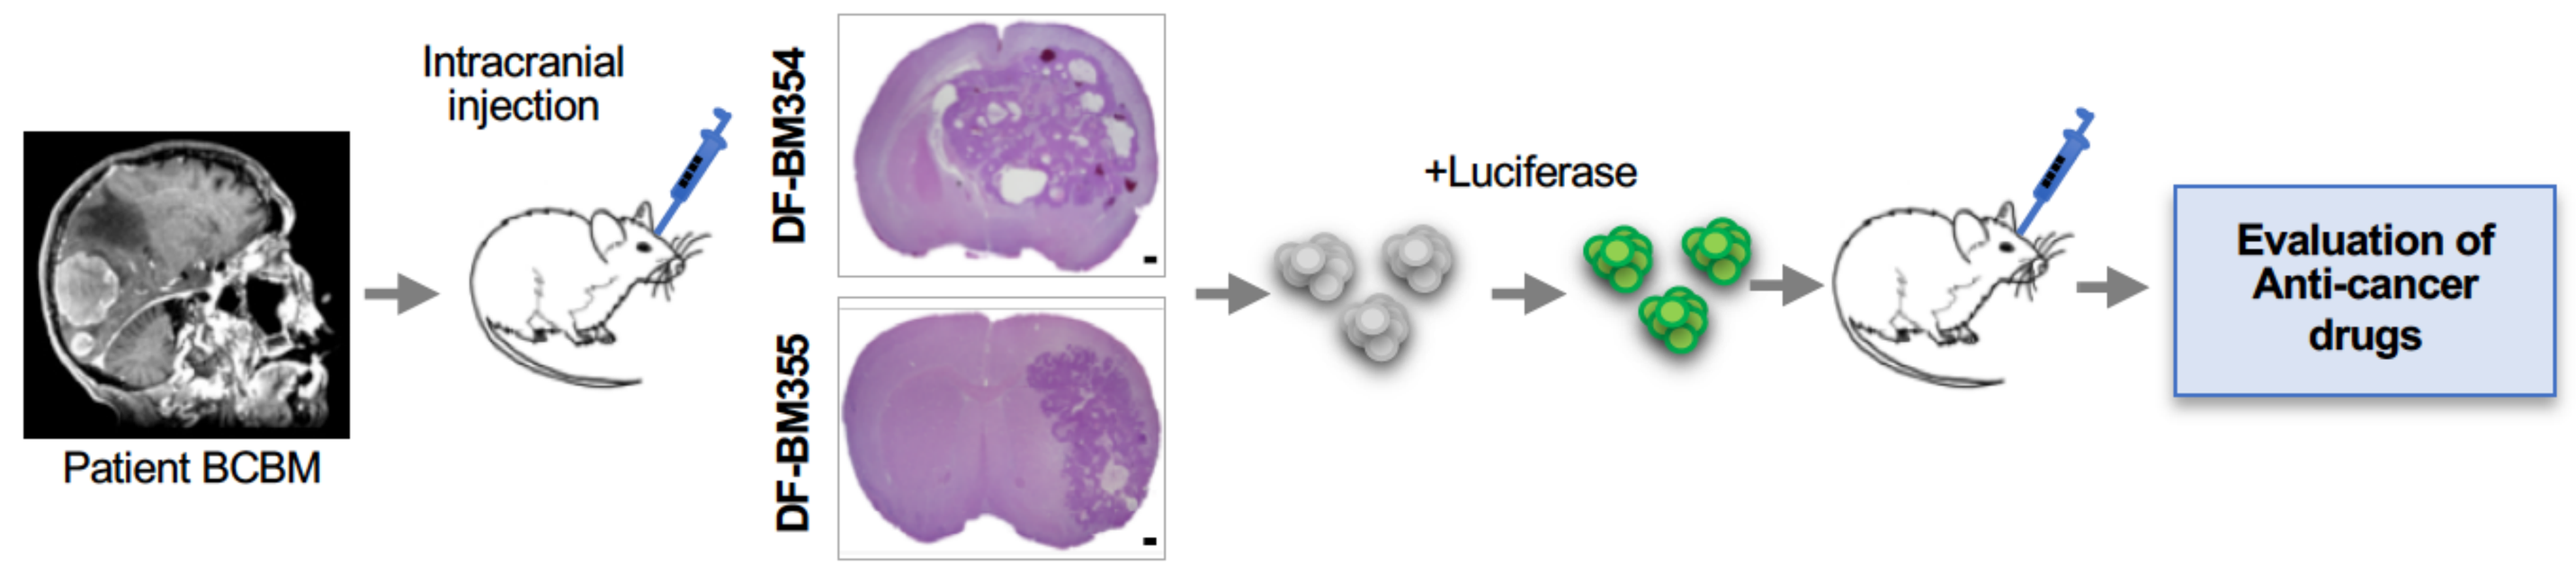
Patient BCBM
Intracranial injection
DF-BM354
DF-BM355
+Luciferase
Evaluation of Anti-cancer drugs

Orthotopic BCBM PDXs resemble the original patient BCBM at the genomic, histopathological, and molecular levels (Ni et al 2016 Nature Medicine).

Combination of neratinib and T-DM1 in DF-BM354 (HER+ER-) model

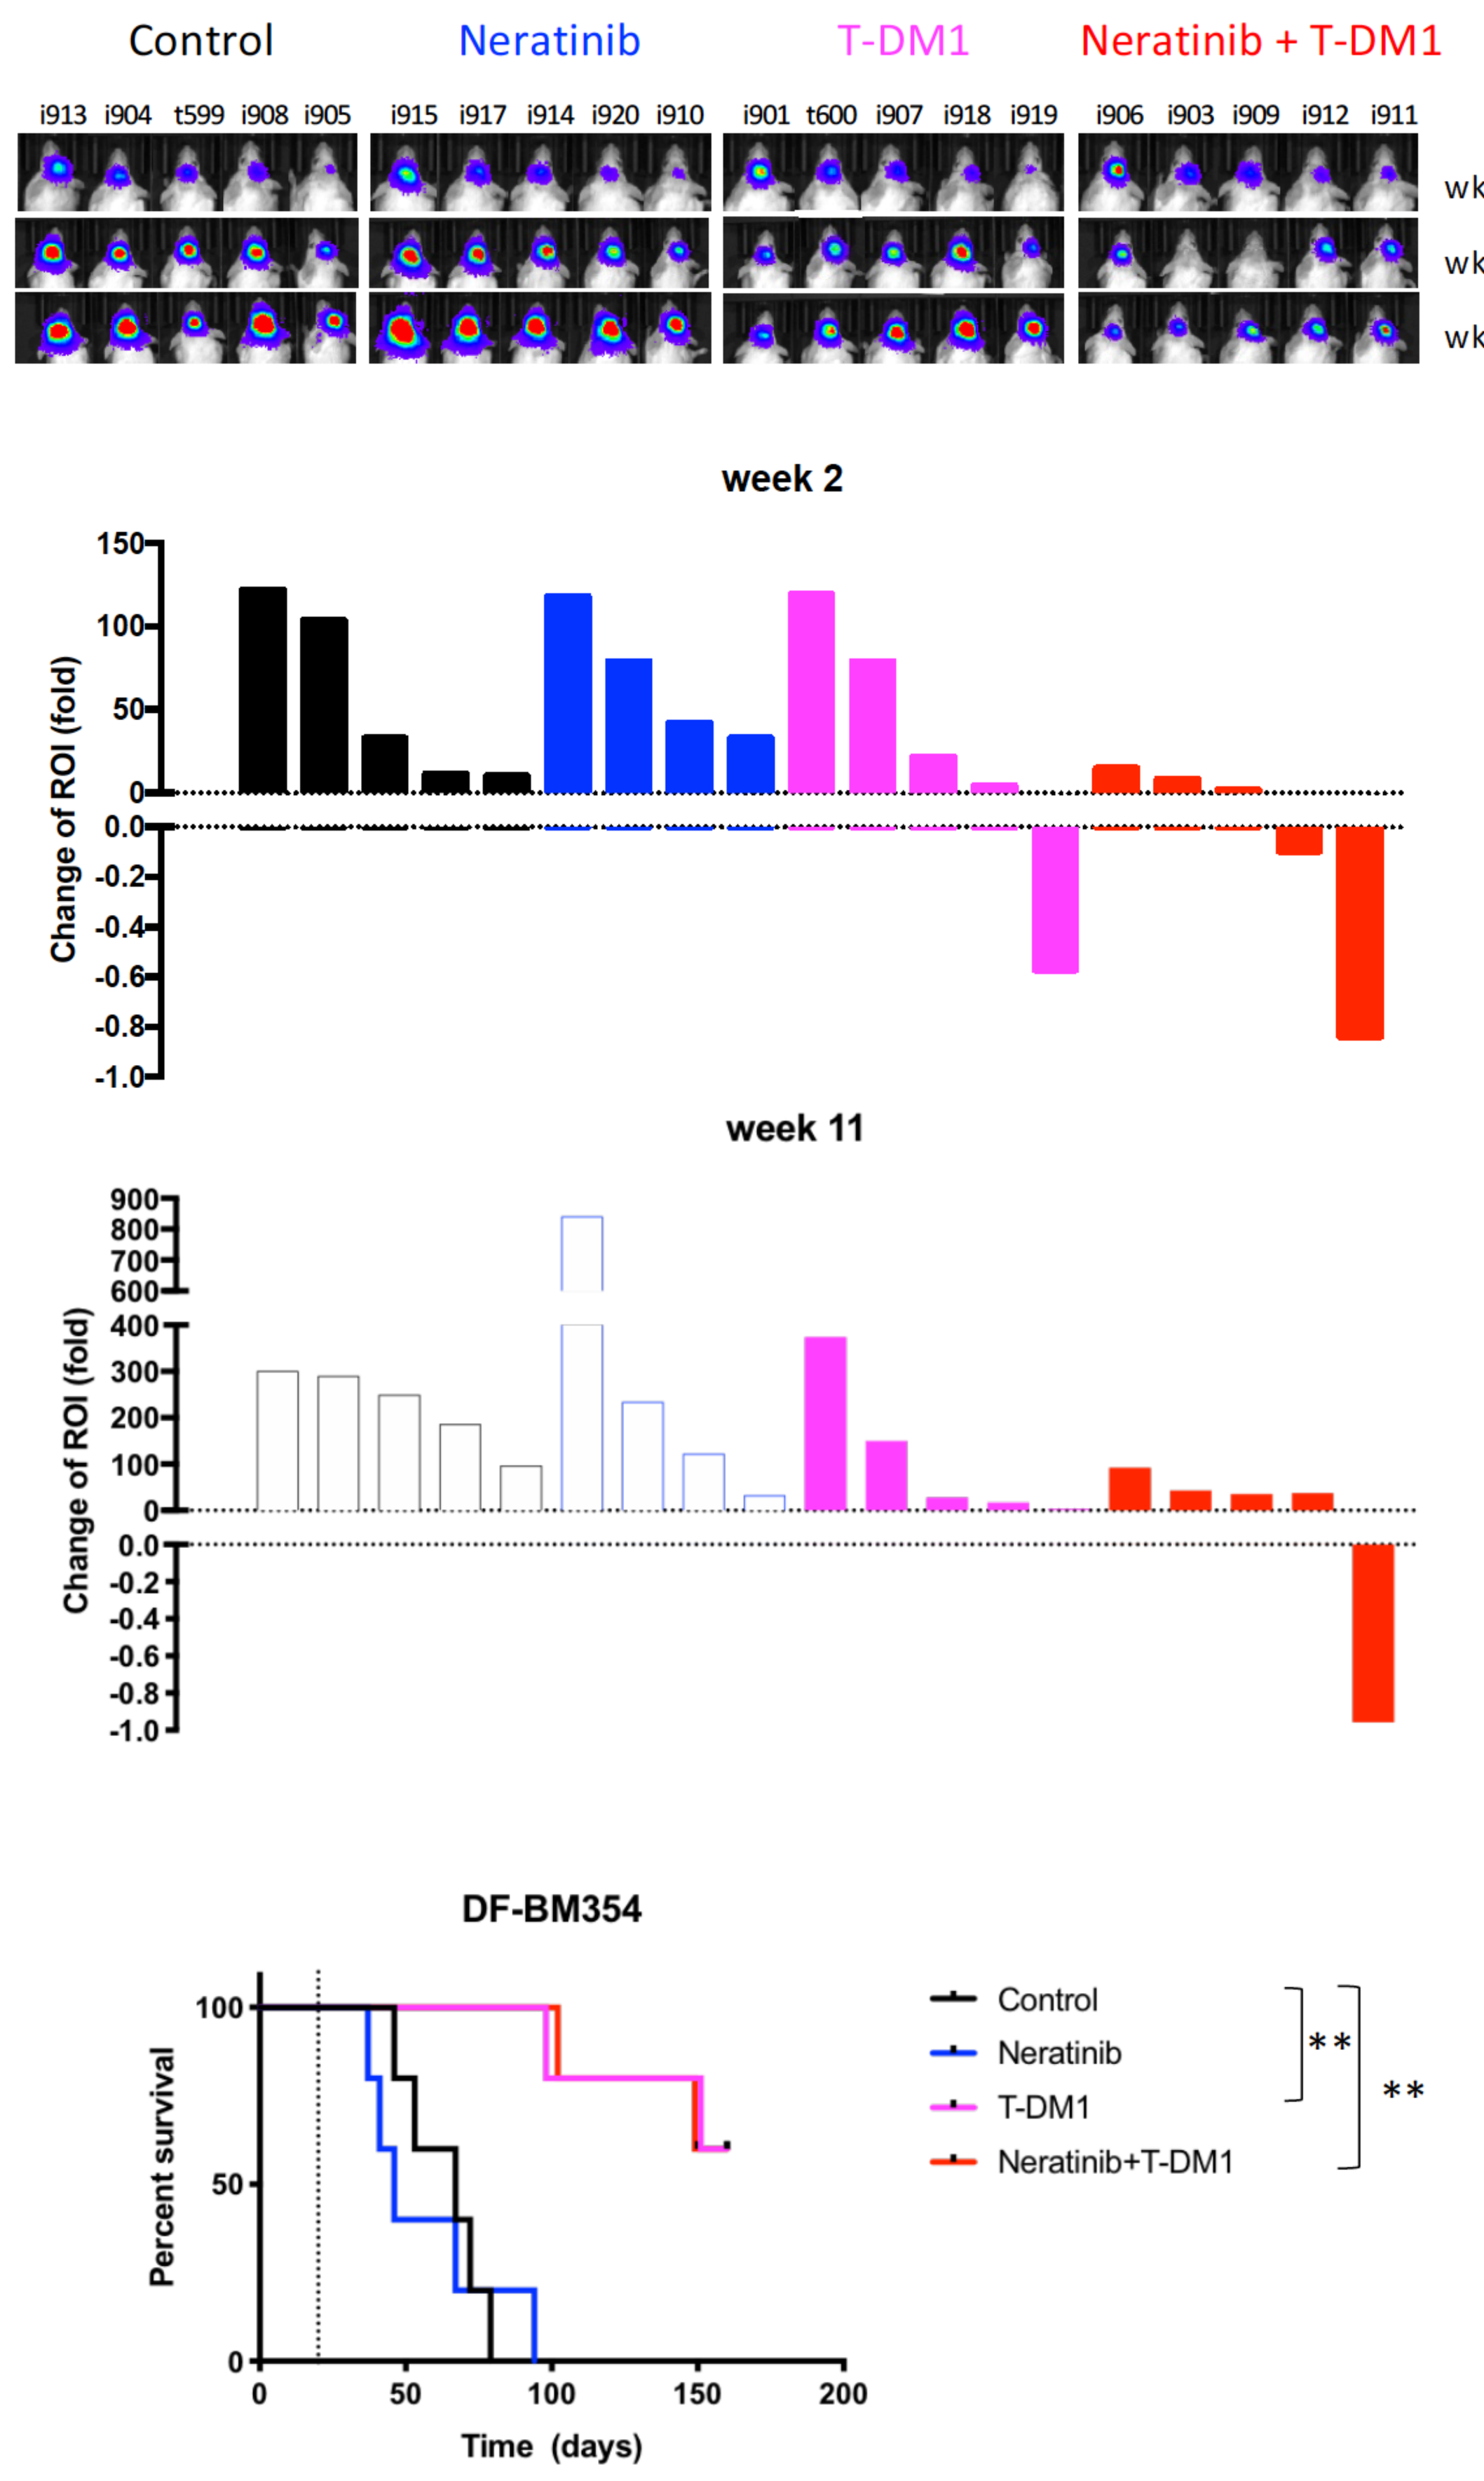
Control
Neratinib
T-DM1
Neratinib + T-DM1
i913 i904 t599 i908 i905
i915 i917 i914 i920 i910
i901 t600 i907 i918 i919
i906 i903 i909 i912 i911
wk0
wk1
wk2
week 2
week 11
Change of ROI (fold)
DF-BM354
Percent survival
Time (days)
Control
Neratinib
T-DM1
Neratinib+T-DM1
**
**

Intracranial DF-BM354 BCBM PDX in mice (1e5 cells / mouse)
Neratinib (40 mg/kg, PO, QD).
T-DM1 (15 mg/kg, IV, Q2W)
Tumor growth monitored by bioluminescent imaging
Log-rank (Mantel-Cox) test (**, $P < 0.01$)

Combination of neratinib and T-DM1 in DF-BM355 (HER2+ER+) model

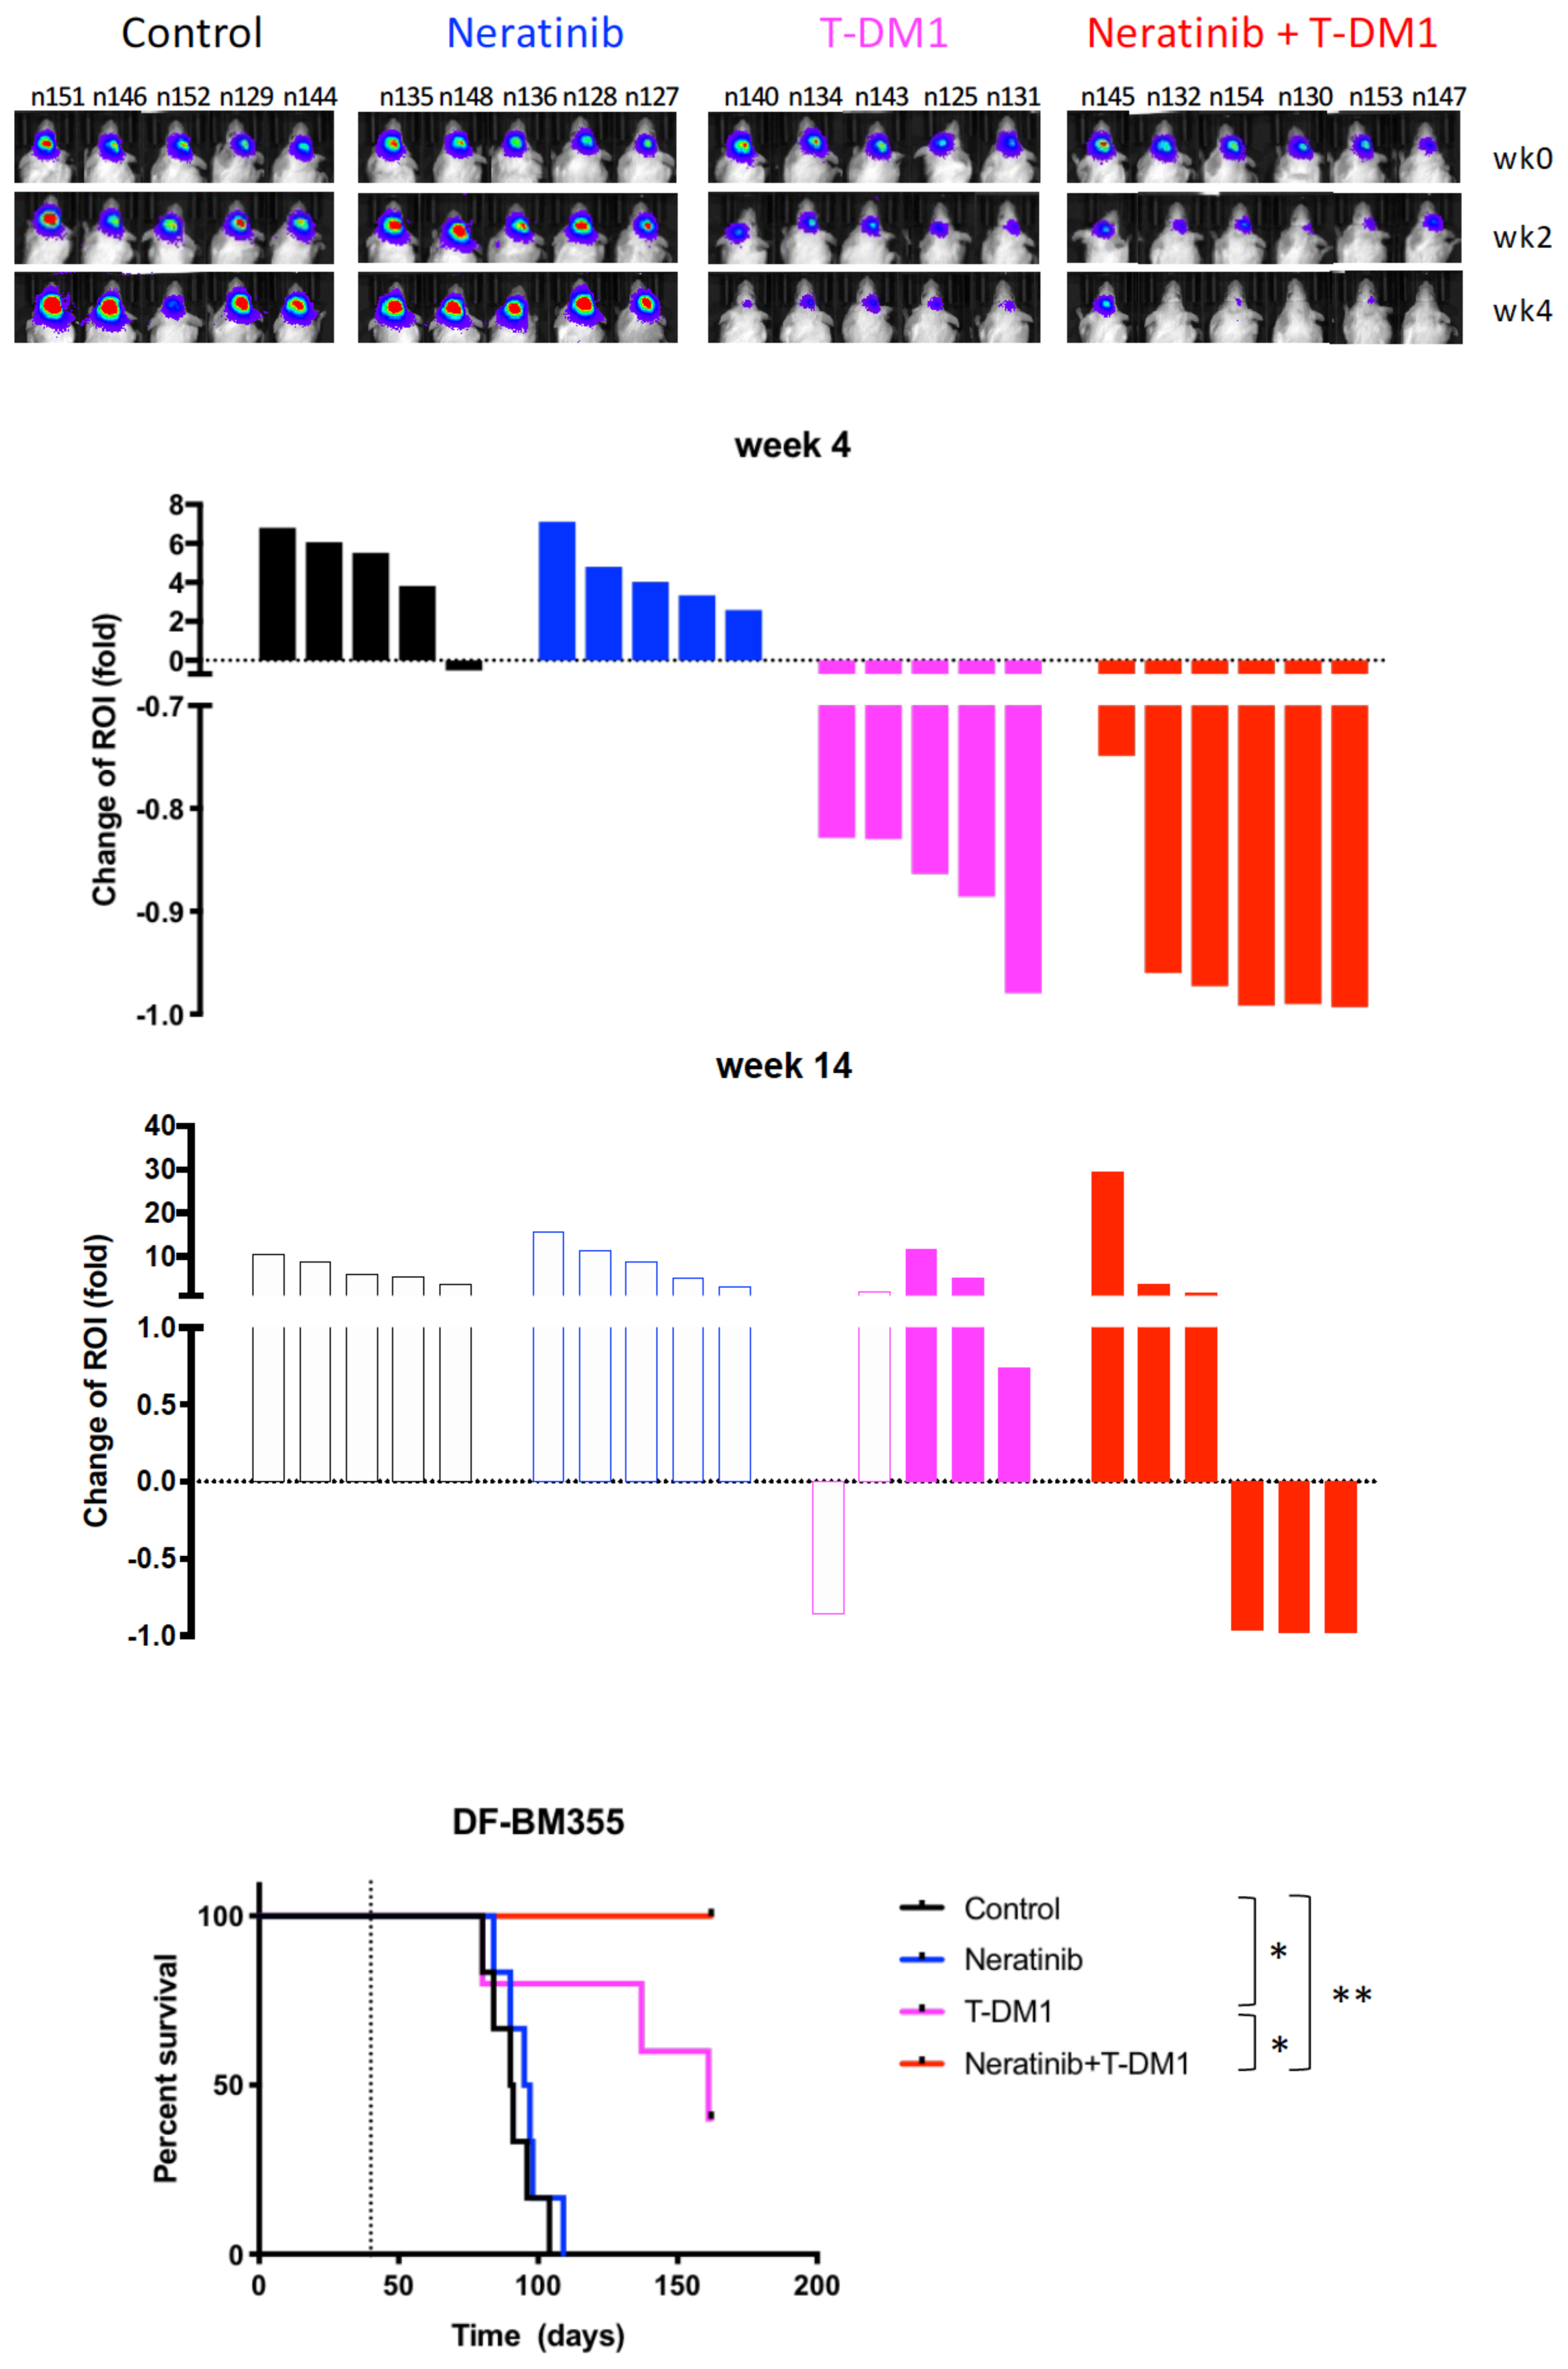
Control
Neratinib
T-DM1
Neratinib + T-DM1
n151 n146 n152 n129 n144
n135 n148 n136 n128 n127
n140 n134 n143 n125 n131
n145 n132 n154 n130 n153 n147
wk0
wk2
wk4
week 4
week 14
Change of ROI (fold)
DF-BM355
Percent survival
Time (days)
Control
Neratinib
T-DM1
Neratinib+T-DM1
*
*
**

Intracranial DF-BM355 BCBM PDX in mice (1e5 cells / mouse)
Neratinib (40 mg/kg, PO, QD).
T-DM1 (15 mg/kg, IV, Q3W)
Tumor growth monitored by bioluminescent imaging
Log-rank (Mantel-Cox) test (*, $P< 0.05$; **, $P < 0.01$)

Neratinib reduces p-HER2 levels in HER2+ BCBM PDX in mouse brain

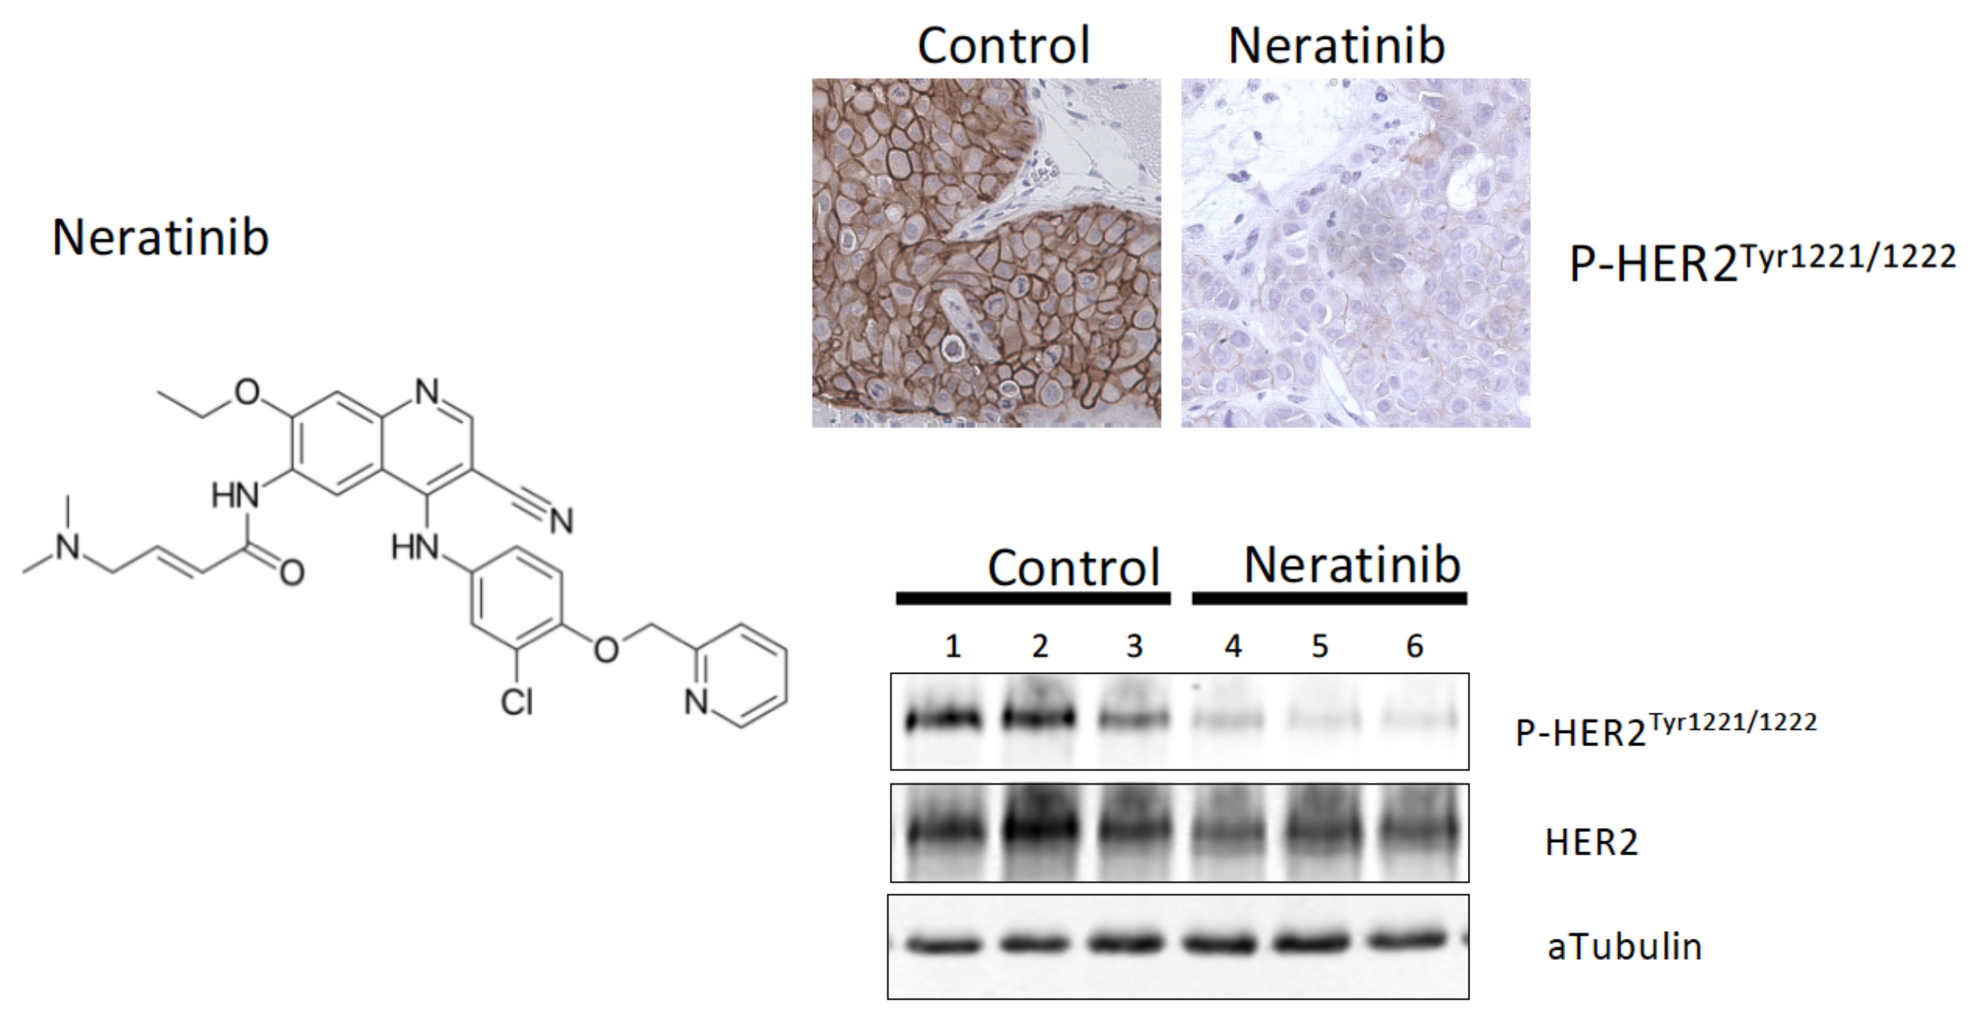
Neratinib
Control
Neratinib
P-HER2 Tyr1221/1222
1 2 3 4 5 6
HER2
aTubulin

Intracranial DF-BM355 BCBM PDX in mice
Neratinib (80 mg/kg, PO, QD x 4).
Tumor collected at 2 hr after last dose

Conclusion

- Neratinib reduced HER2 phosphorylation in the orthotopic HER2-positive BCBM PDX tumors, suggesting that it can cross the blood brain barrier.
- Neratinib as a single agent could not inhibit the growth of orthotopic HER2-positive BCBM PDX tumors.
- T-DM1 alone can inhibit BCBM PDX tumor growth but the effect is not sustained.
- The combination of neratinib and T-DM1 demonstrated efficacy beyond the single agent treatment of HER2-positive brain metastatic breast cancer.
- Further work will elucidate drivers of resistance and susceptibility to HER2-inhibitors in HER2-positive BCBM models.
- This preclinical study further supports the launch of a clinical trial (NCT01494662) to test the combination of neratinib and T-DM1 in HER2-positive BCBM patients.

Acknowledgements

NIH/NCI; The Stand Up to Cancer; Breast Cancer Research Foundation; PUMA biotechnology